Verification of a Complete Sample to Insight® Liquid Biopsy Workflow – NGS of ccfDNA From Stabilized Blood

Tomasz Krenz, Andrea Ullius, Ricardo Huebel, Thorsten Voss, Eric Provencher, Timothy R. Buirkle and Daniel Groelz
PreAnalytiX GmbH, Hombrechtikon, Switzerland

PreAnalytiX
A QIAGEN / BD Company

AACR Annual Meeting 2018
Poster number 4592

Introduction

Analysis of circulating cell-free DNA (ccfDNA) from blood in the fields of life science, clinical research and beyond is expanding and has become widely accepted. The complete workflow entails collection and stabilization of blood, transport, plasma processing, extraction of ccfDNA and downstream analysis. Considering the usual low sample input and required high sensitivity for quality control (QC), optimal outcomes require verification of the entire workflow.

In this verification study, human blood was collected, stabilized, and then processed to isolate ccfDNA from plasma using the PAXgene® Blood ccfDNA System for subsequent next-generation sequencing (NGS) on the QIAGEN GeneReader® system. To verify the workflow, several sample QC criteria were assessed at each step of the workflow, including ccfDNA yield, target enrichment, library preparation, target sequencing and analysis.

Methods

Blood from 60 healthy consented donors was collected into PAXgene Blood ccfDNA Tubes* and stored for 7 days at 25°C to simulate a typical processing delay in a routine setting. Automated ccfDNA extraction was performed on the QIAsymphony® SP instrument using the QIAsymphony PAXgene Blood ccfDNA Kit* and protocol. ccfDNA stability after blood storage was confirmed by qPCR. ccfDNA samples were sequenced on the QIAGEN GeneReader NGS System, including the GeneRead™ QIAact Actionable Insights Tumor (AIT) Panel* for PCR target enrichment, library preparation on the QIAcube® instrument, QC with capillary electrophoresis, sequencing on the GeneReader instrument* and data management with the QIAGEN Clinical Insight (QCI™) Analyze* tool.

Target Enrichment & Library Preparation

- All samples (60/60) passed the required QC criteria after target enrichment (amplicon size <170 bp) and library preparation (increase of amplicon size by 92 bp (adapters) and absence of unspecific products).

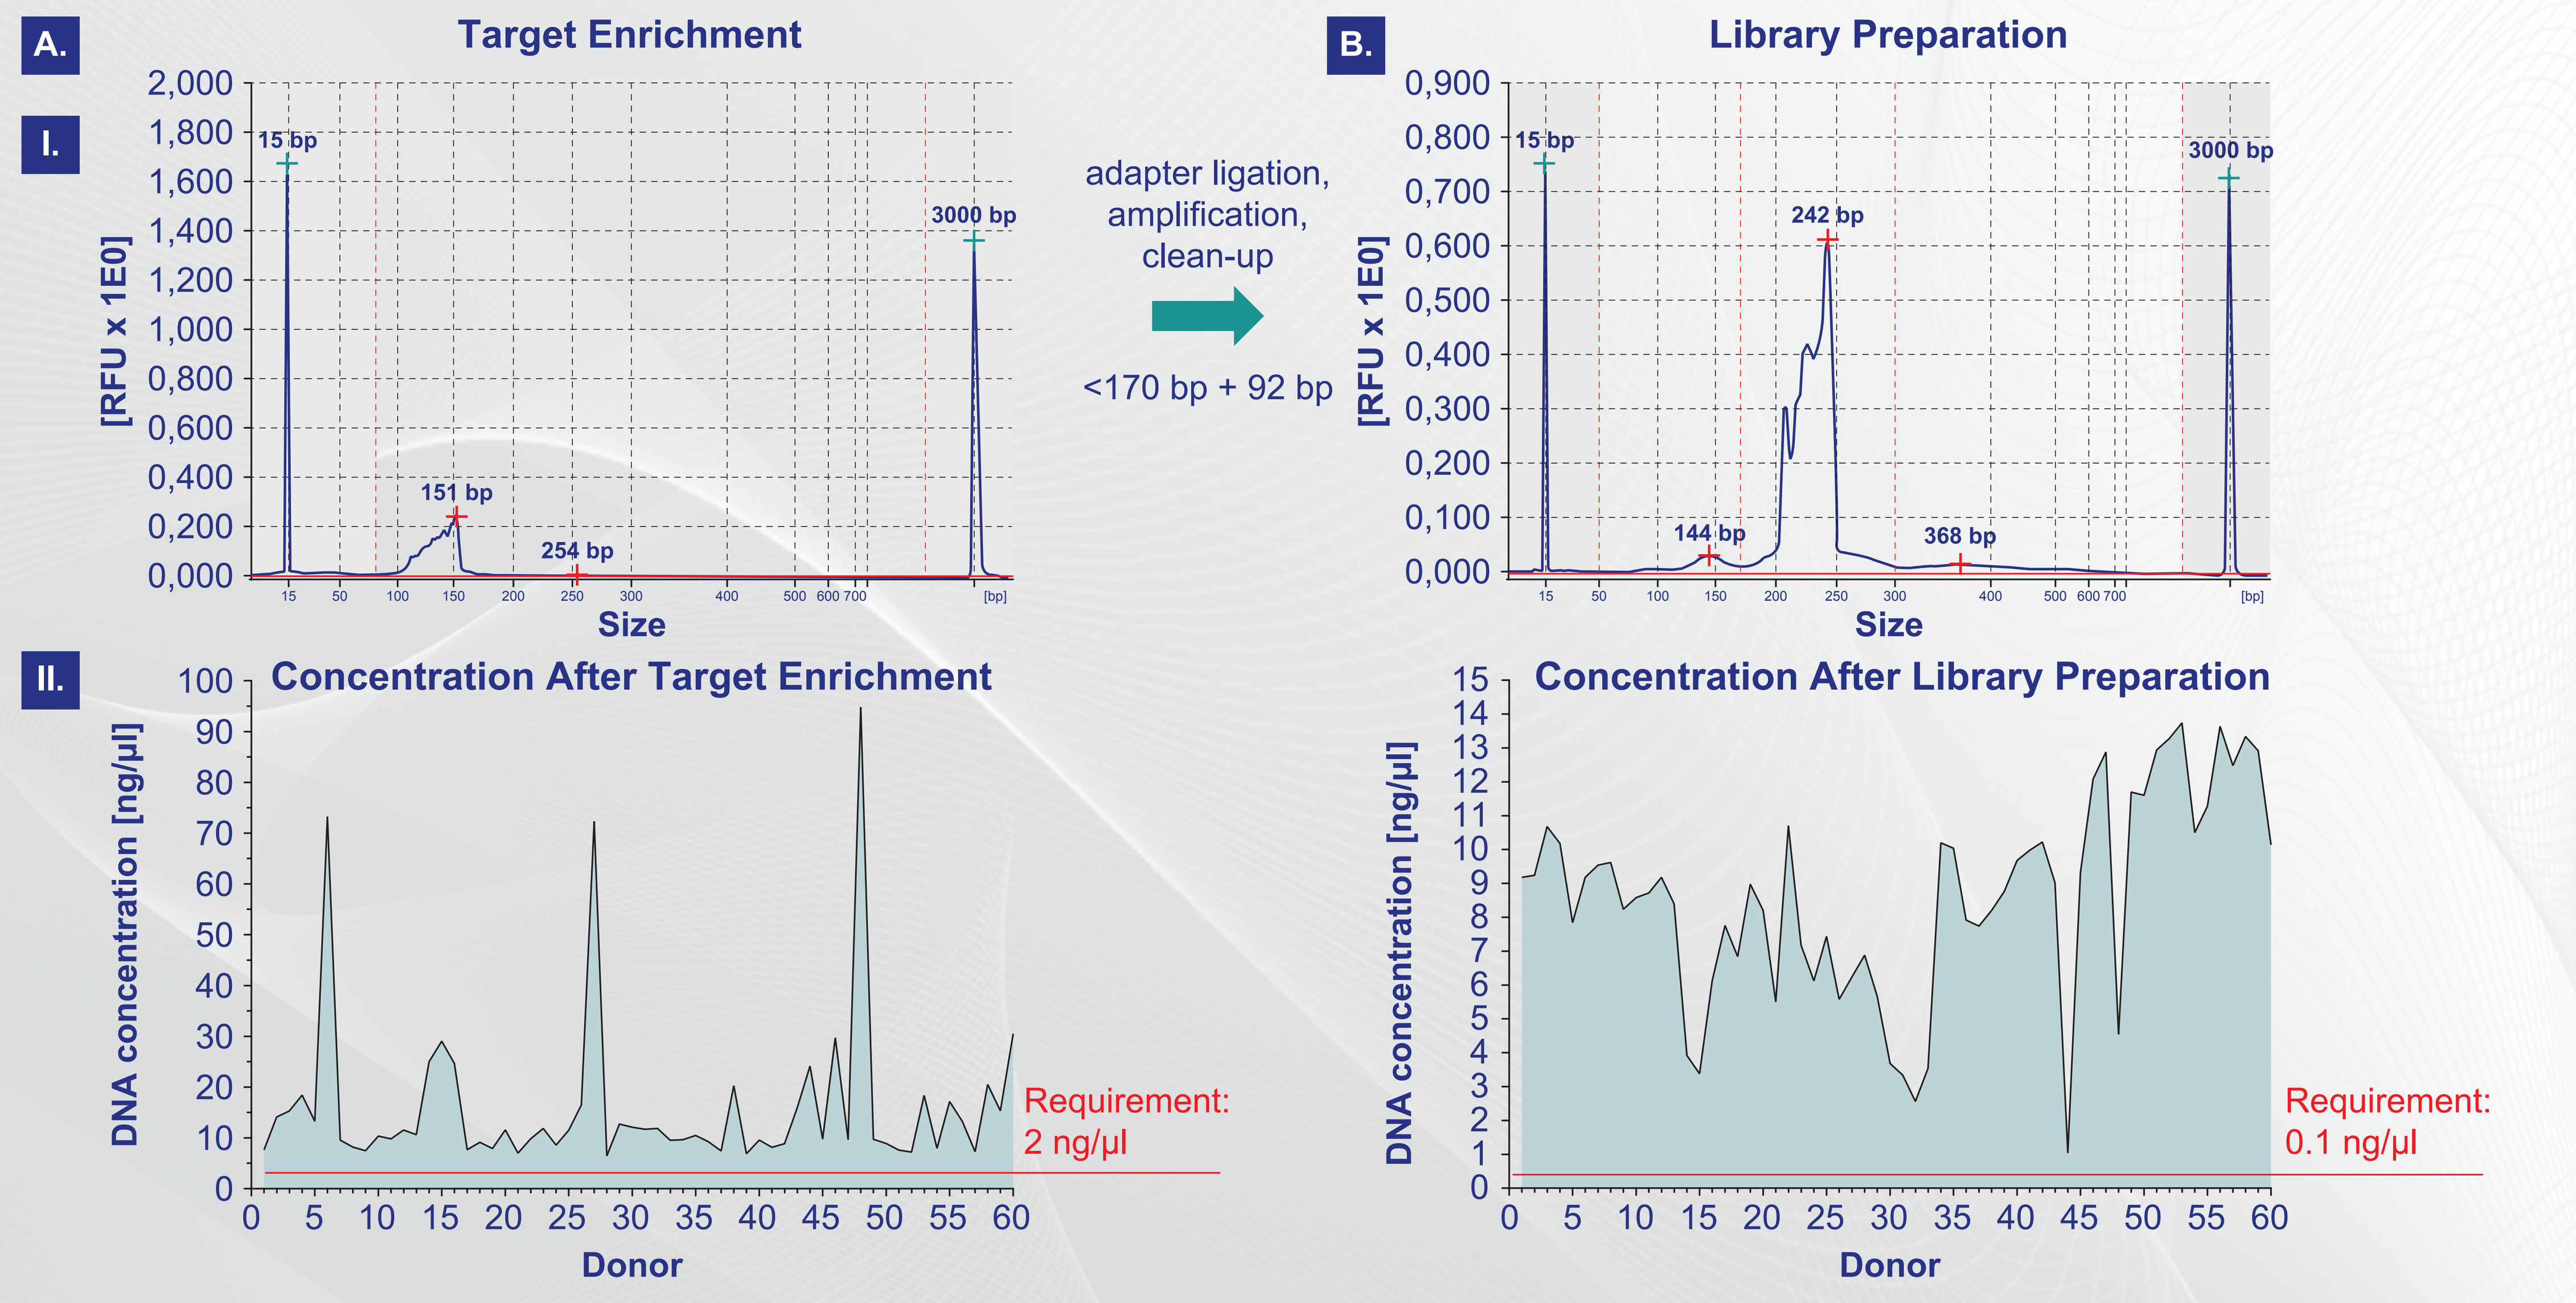
A.
Target Enrichment
I.
[RFU x 1E0]
15 bp
151 bp
254 bp
3000 bp
Size
adapter ligation, amplification, clean-up
<170 bp + 92 bp
B.
Library Preparation
15 bp
144 bp
242 bp
368 bp
3000 bp
Size
II.
Concentration After Target Enrichment
DNA concentration [ng/µl]
Requirement: 2 ng/µl
Donor
Concentration After Library Preparation
Requirement: 0.1 ng/µl
Donor

Compatibility of the liquid biopsy workflow in target enrichment (A) and library preparation (B) NGS procedures. Blood from 60 healthy donors was stored in PAXgene Blood ccfDNA Tubes for 7 days at 25°C prior to ccfDNA extraction. Maximum volume of ccfDNA eluate was used for the upfront NGS workflow using the GeneReader workflow and the GeneRead QIAact Actionable Insights Tumor (AIT) panel. The products after target enrichment (A) and library preparation (B) were analyzed for correct fragment size (I.) and total product concentration (II.) using capillary electrophoresis on the QIAxcel instrument. Representative electropherograms and single data points are shown. The red line indicates the minimal DNA concentration requirement for the next NGS step.

Study Design – ccfDNA NGS Workflow

- Blood from 60 healthy donors
- PAXgene Blood ccfDNA Tube, QIAsymphony PAXgene Blood ccfDNA Kit, GeneReader NGS system: Sample to Insight

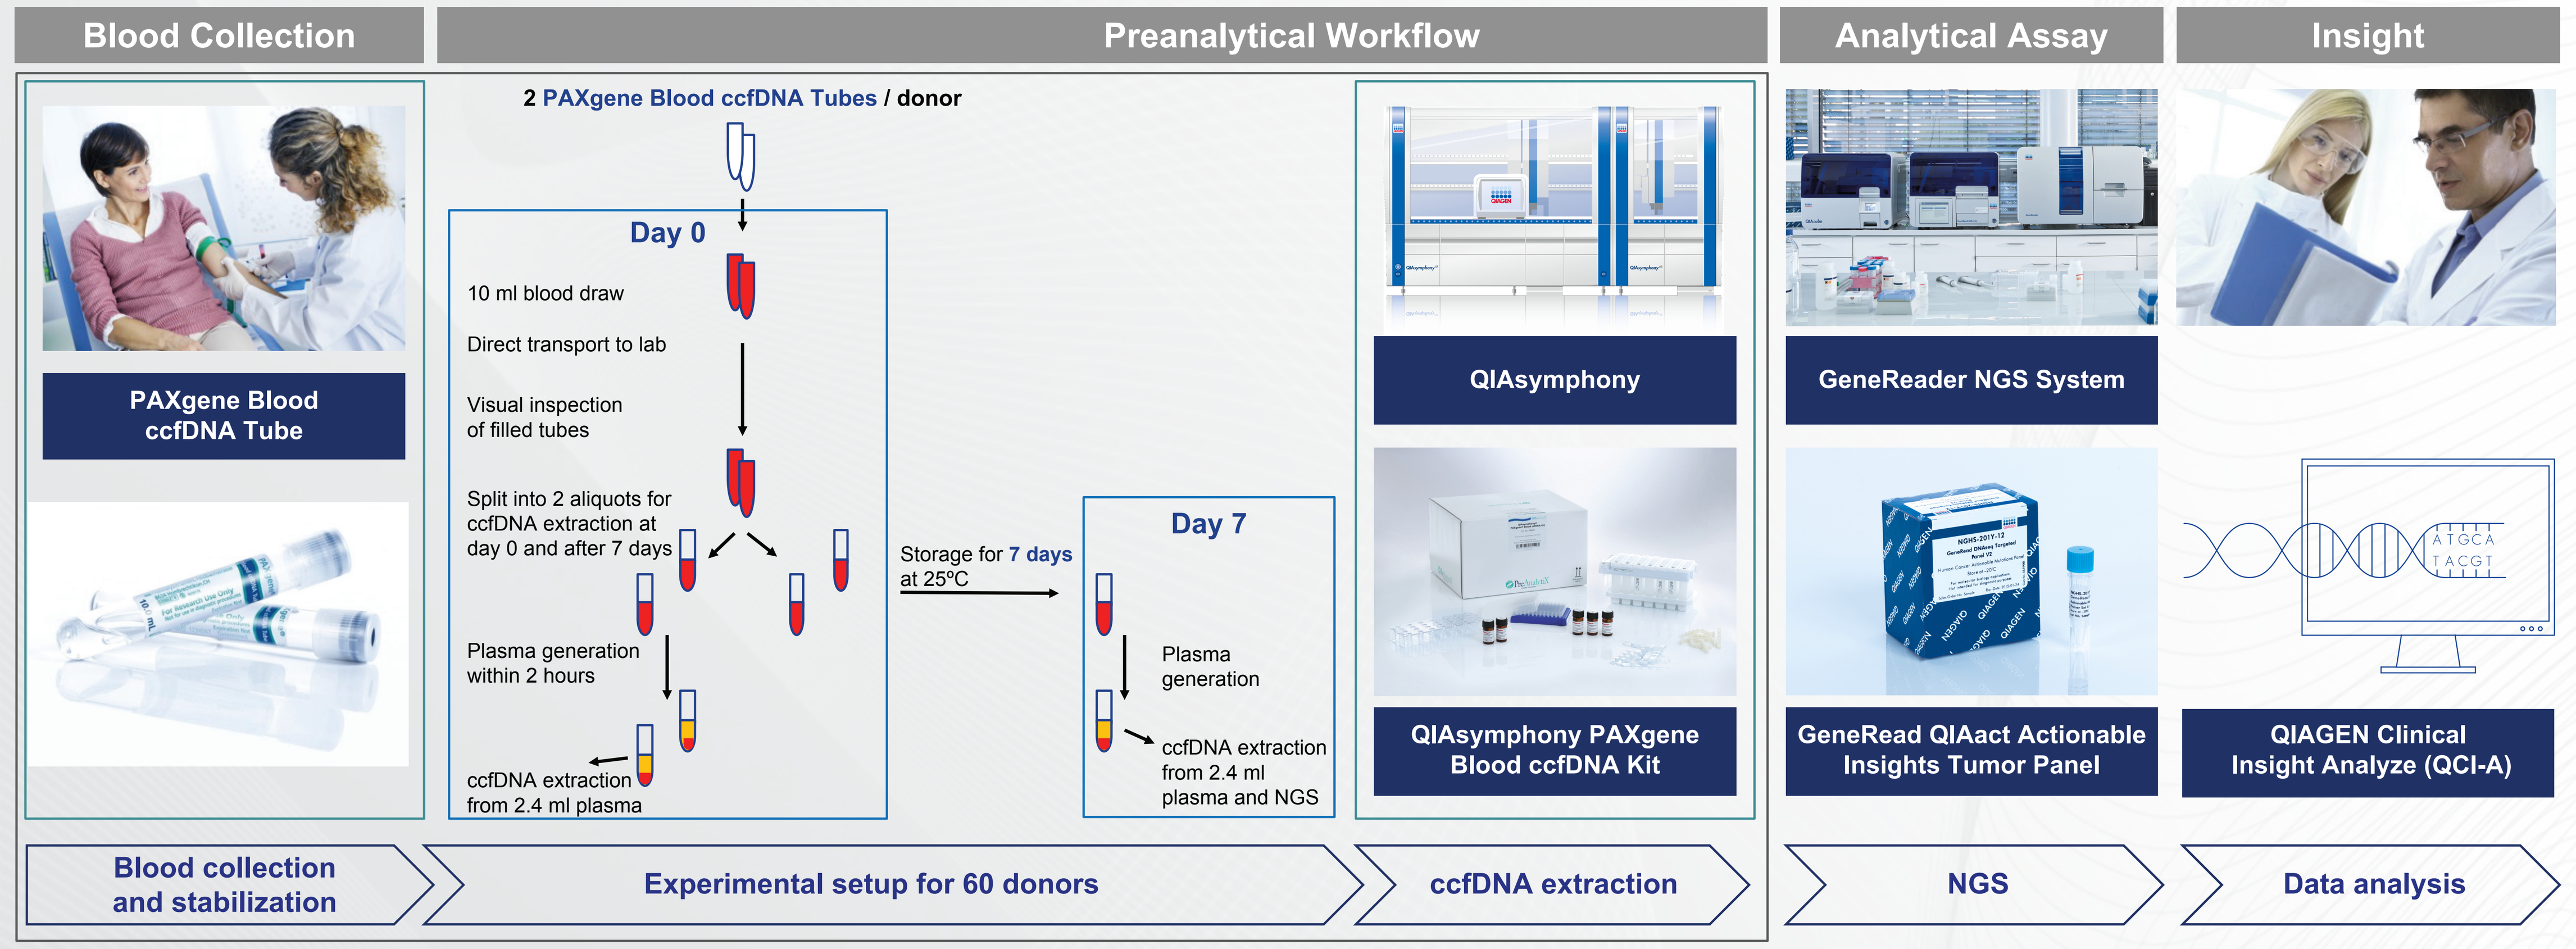
Blood Collection
Preanalytical Workflow
Analytical Assay
Insight
PAXgene Blood ccfDNA Tube
2 PAXgene Blood ccfDNA Tubes / donor
Day 0
10 ml blood draw
Direct transport to lab
Visual inspection of filled tubes
Split into 2 aliquots for ccfDNA extraction at day 0 and after 7 days
Plasma generation within 2 hours
ccfDNA extraction from 2.4 ml plasma
Storage for 7 days at 25°C
Day 7
Plasma generation
ccfDNA extraction from 2.4 ml plasma and NGS
QIAsymphony
QIAsymphony PAXgene Blood ccfDNA Kit
GeneReader NGS System
GeneRead QIAact Actionable Insights Tumor Panel
QIAGEN Clinical Insight Analyze (QCI-A)
Blood collection and stabilization
Experimental setup for 60 donors
ccfDNA extraction
NGS
Data analysis

In Situ ccfDNA Stability

- After blood storage for 7 days, ccfDNA yield was comparable to ccfDNA yield observed directly after blood draw.

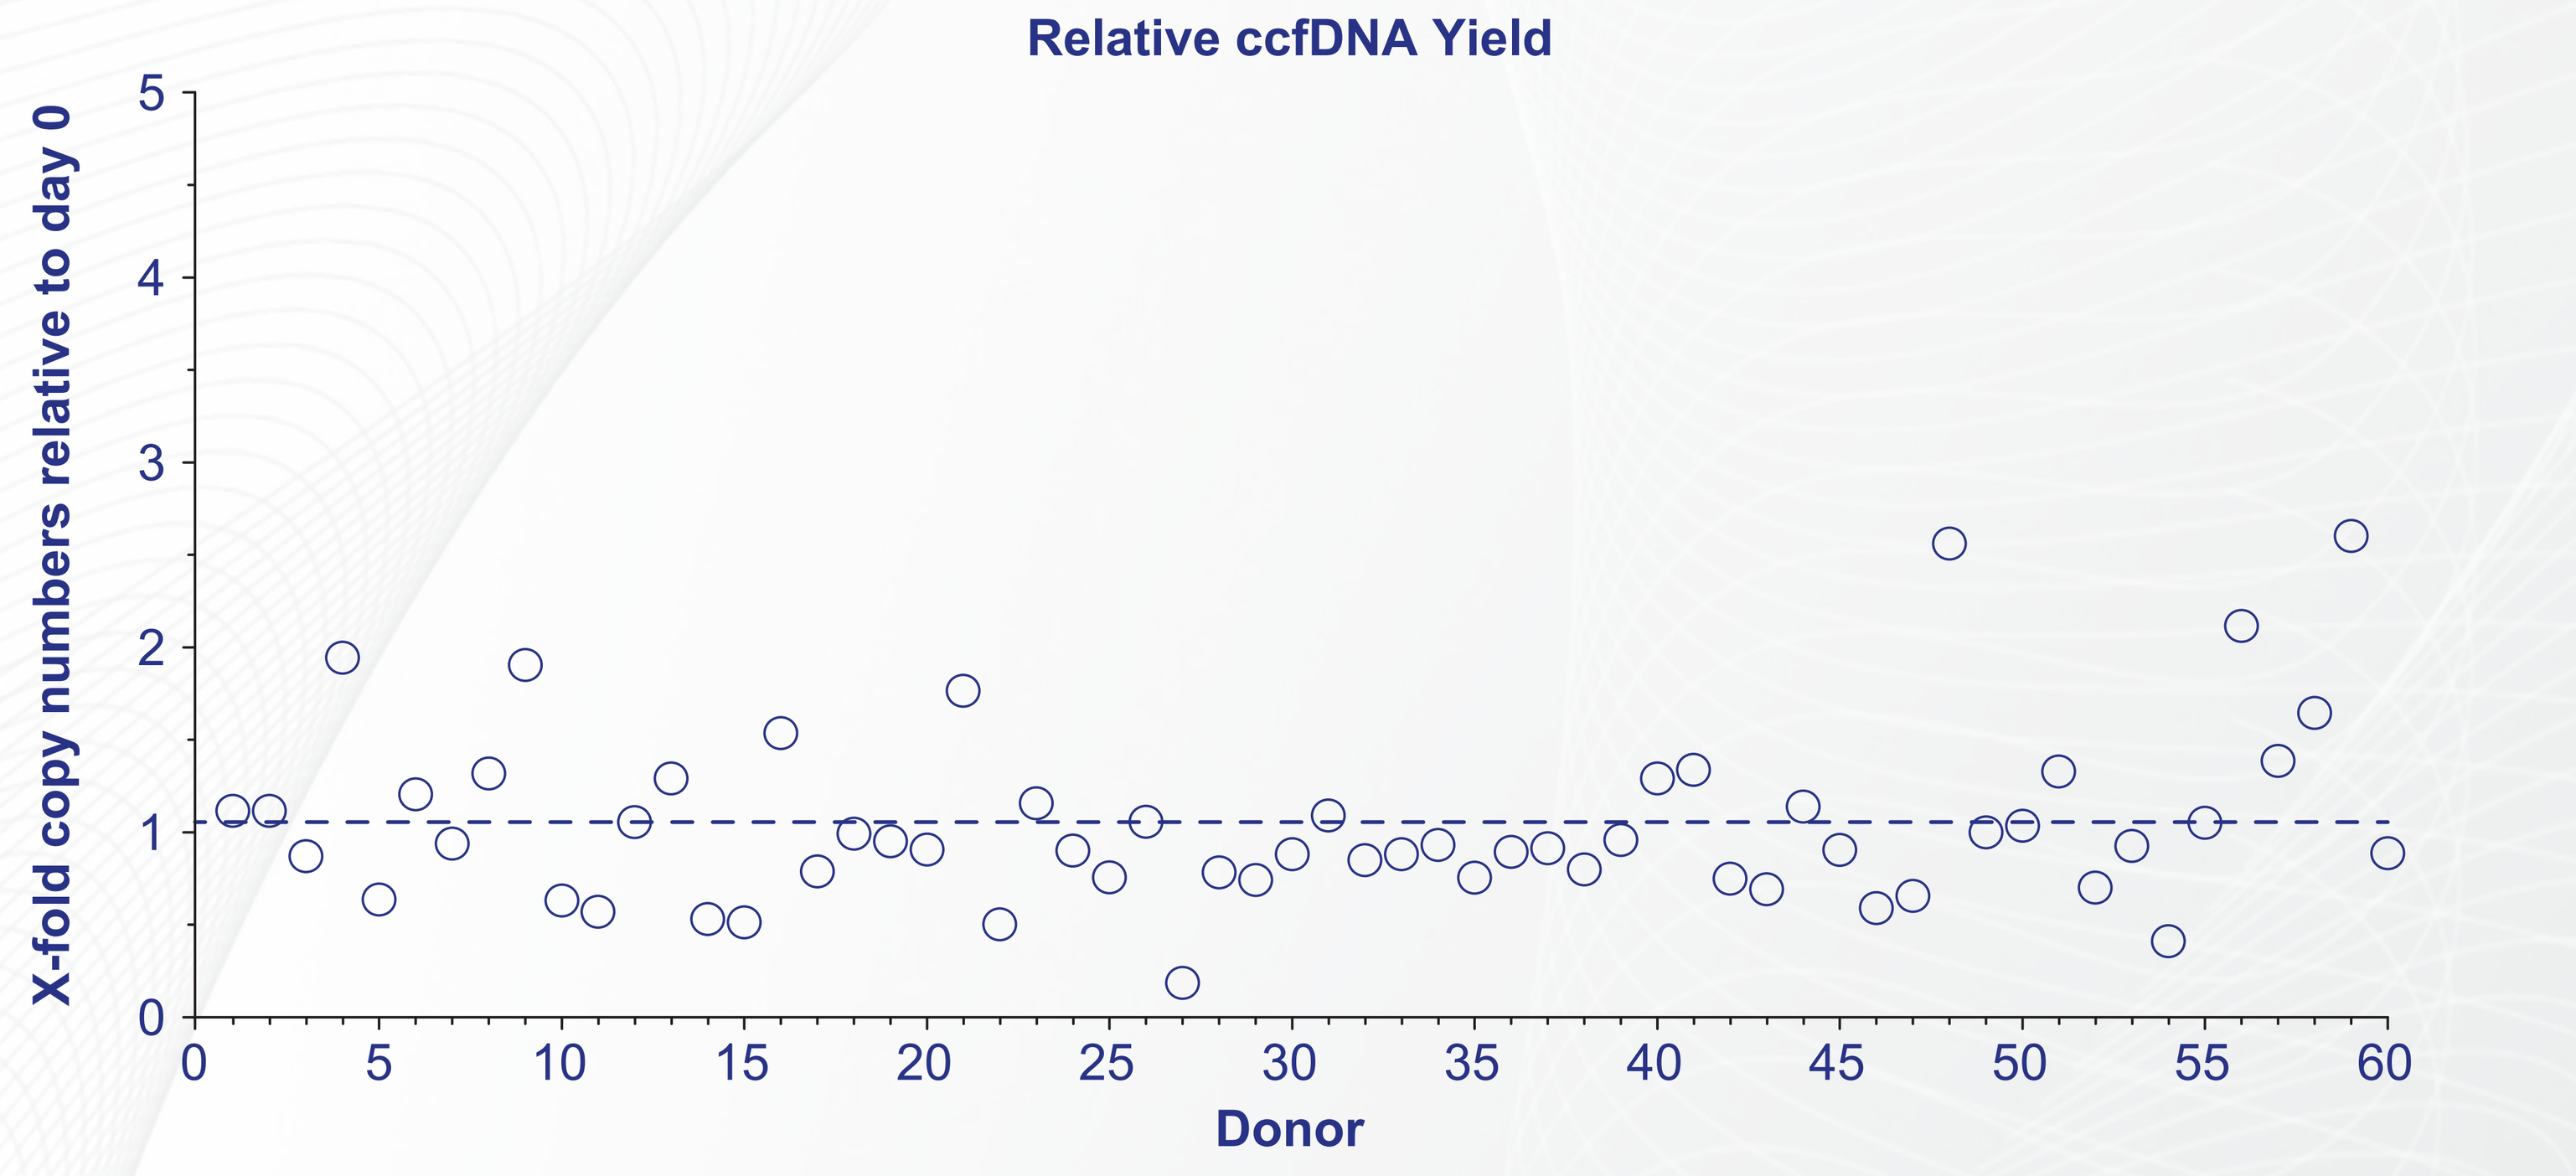
Relative ccfDNA Yield
X-fold copy numbers relative to day 0
Donor

Relative copy number change of 18S rDNA 66 bp fragments in PAXgene Blood ccfDNA Tube. Copy numbers from 60 healthy blood donors were determined with a Primer/Probe qPCR assay directly after blood collection (day 0) and after blood storage for 7 days at 25°C (day 7). Single data points are shown for each donor. The mean value (mean = 1.045) is shown as a blue dotted line. Ideal ratio would be 1.0 meaning no change versus day 0. All values are within the accepted range.

GeneReader NGS Results: QC and Variant Analysis

- All GeneReader NGS acceptance criteria were met for every sample, including region of interest with coverage of bases >500x (>90%), region of interest with coverage of bases >200x (>95%) and reads above average quality 25 (>80%).
- Across all samples, 17 ± 5 different variants in 7 ± 1 different genes were identified in healthy donors using the AIT panel.

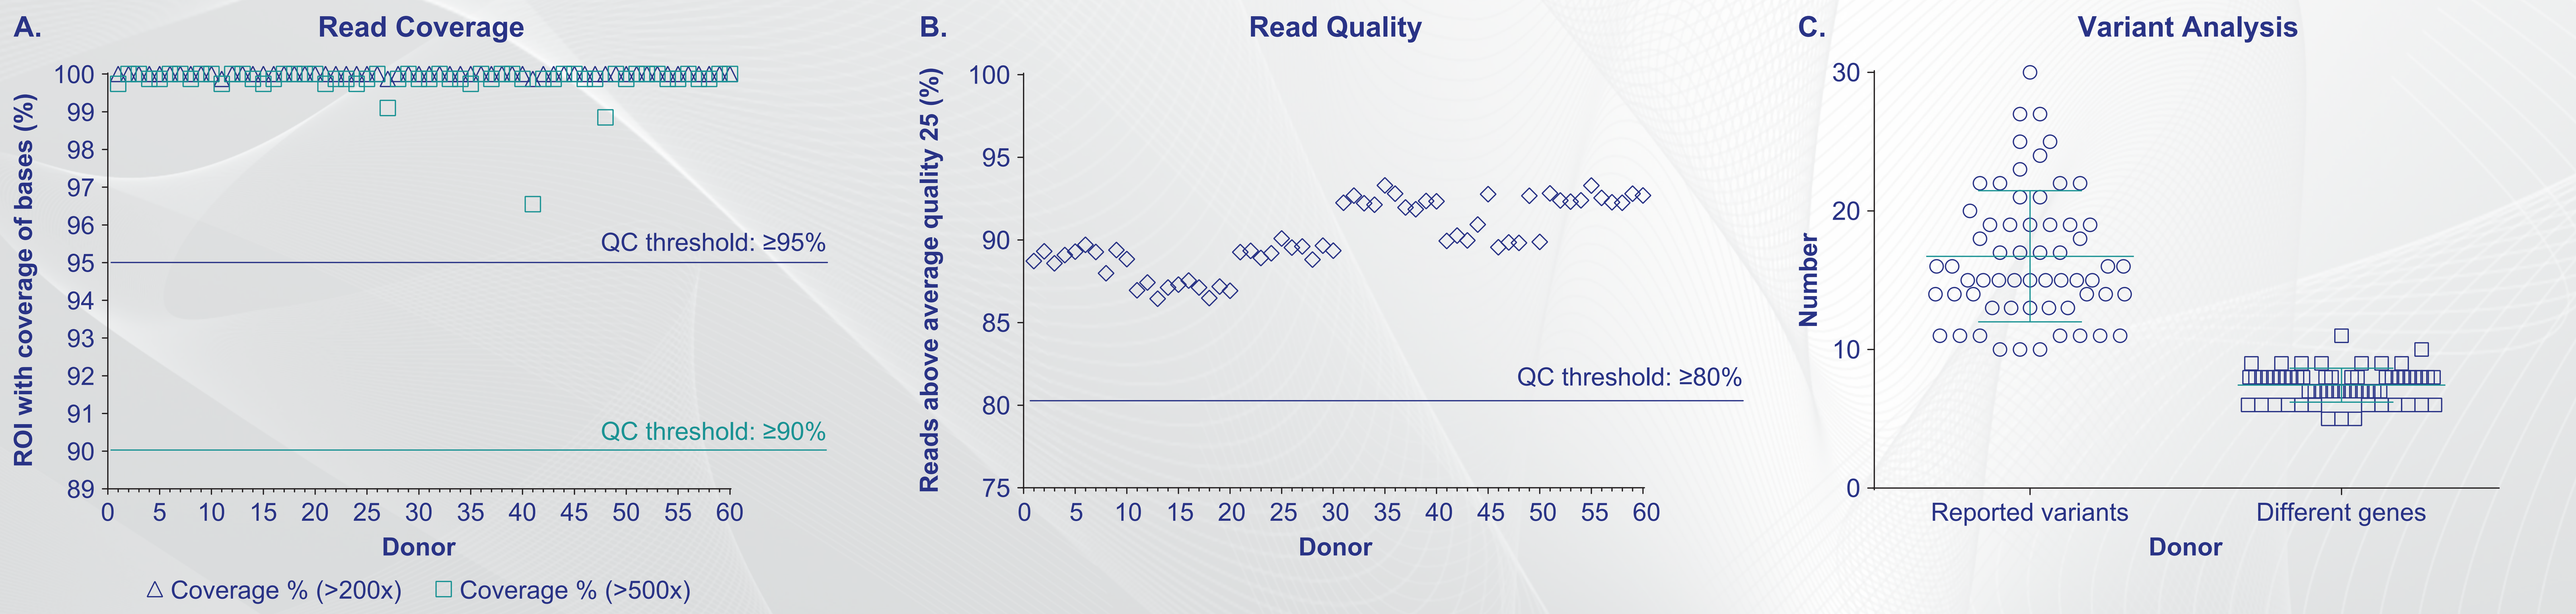
A.
Read Coverage
ROI with coverage of bases (%)
QC threshold: ≥95%
QC threshold: ≥90%
Donor
△ Coverage % (>200x) □ Coverage % (>500x)
B.
Read Quality
Reads above average quality 25 (%)
QC threshold: ≥80%
Donor
C.
Variant Analysis
Number
Reported variants
Different genes
Donor

NGS compatibility of the liquid biopsy workflow. Blood from 60 healthy donors was stored in PAXgene Blood ccfDNA Tubes for 7 days at 25°C prior to ccfDNA extraction. NGS was performed on the GeneReader system using the GeneRead QIAact Actionable Insights Tumor (AIT) panel. Sequencing data was analyzed with the QIAGEN Clinical Insight Analyze (QCI-A) tool for quality control (QC) parameters (A, B) and gene variant detection (C). System QC thresholds for 3 of the most important NGS QC parameters (coverage 500x, coverage 200x, reads above average quality 25) define "passed" or "out-of-spec" samples. Variant analysis was carried out for every individual sample and all gene variants of the panel. The AIT panel contains 773 variant positions in 12 genes.

Conclusion

- NGS is one of the most important applications for analysis of ccfDNA in research and clinical settings. Stabilization of whole blood for sensitive ctDNA applications such as NGS is mandatory.
- ccfDNA yield in PAXgene Blood ccfDNA Tubes is stable for 7 days at room temperature.
- Quality control acceptance criteria for the GeneReader NGS system were met for 100% of analyzed ccfDNA samples.
- ccfDNA stabilized and extracted with the PAXgene Blood ccfDNA System is highly suitable for NGS applications.

Disclaimer: *For Research Use Only. Not for use in diagnostic procedures.

For up-to-date licensing information and product-specific disclaimers, see the respective QIAGEN and PreAnalytiX kit handbook or user manual.

Trademarks are the property of their respective owners.

© 2018 PreAnalytiX GmbH. PreAnalytiX, the PreAnalytiX Logo and all other trademarks are property of PreAnalytiX GmbH, Hombrechtikon, CH.

BLOOD · TISSUE · BONE MARROW
The better the source, the more to eXplore.